Patient journey, healthcare resource utilization, and mortality in IBM: A US-based claims analysis

Raymond E. Mankoski,[1] Karen Tubridy,[1] Jacob Winthrop,[2] Chris Koch,[2] Elie Naddaf[3]

[1]Abcuro Inc., Newton, MA, USA; [2]Real Chemistry, New York, NY, USA; [3]Department of Neurology, Mayo Clinic, Rochester, MN, USA

Introduction

- Inclusion body myositis (IBM) is a rare, progressive inflammatory myopathy in which highly differentiated, cytotoxic CD8+ T cells expressing killer cell lectin-like receptor G1 (KLRG1) invade healthy skeletal muscle tissue[1-3]
 - IBM generally presents in adults aged >40 years, and clinical hallmarks include asymmetric weakness and dysphagia that lead to severe disability over time[1,4,5]
- Studies evaluating diagnostic journey, associated comorbidities, healthcare resource utilization (HCRU), and mortality in patients with IBM to date have been limited in patient population size[5-8]

Objective

- To evaluate the patient journey, comorbidities, HCRU, and mortality of patients with IBM using a large US claims database

Methods

Claims analyses

- The Real Chemistry US claims database was used to identify patients with ≥2 IBM diagnosis claims based on the International Classification of Diseases, Tenth Revision (ICD-10) classification system (ICD-10 code G72.41) and an age- and sex-matched control cohort without IBM (**Figure 1**)
 - For patients with IBM, the first IBM diagnosis claim had to occur between 1/1/2020 and 7/30/2025
- Patient characteristics, time to diagnosis, comorbidities, HCRU, and mortality were assessed
- Mortality was evaluated using standardized mortality ratios (SMR) expressed as: Total yearly IBM deaths/US population mortality rate

Figure 1. IBM patient cohort of interest

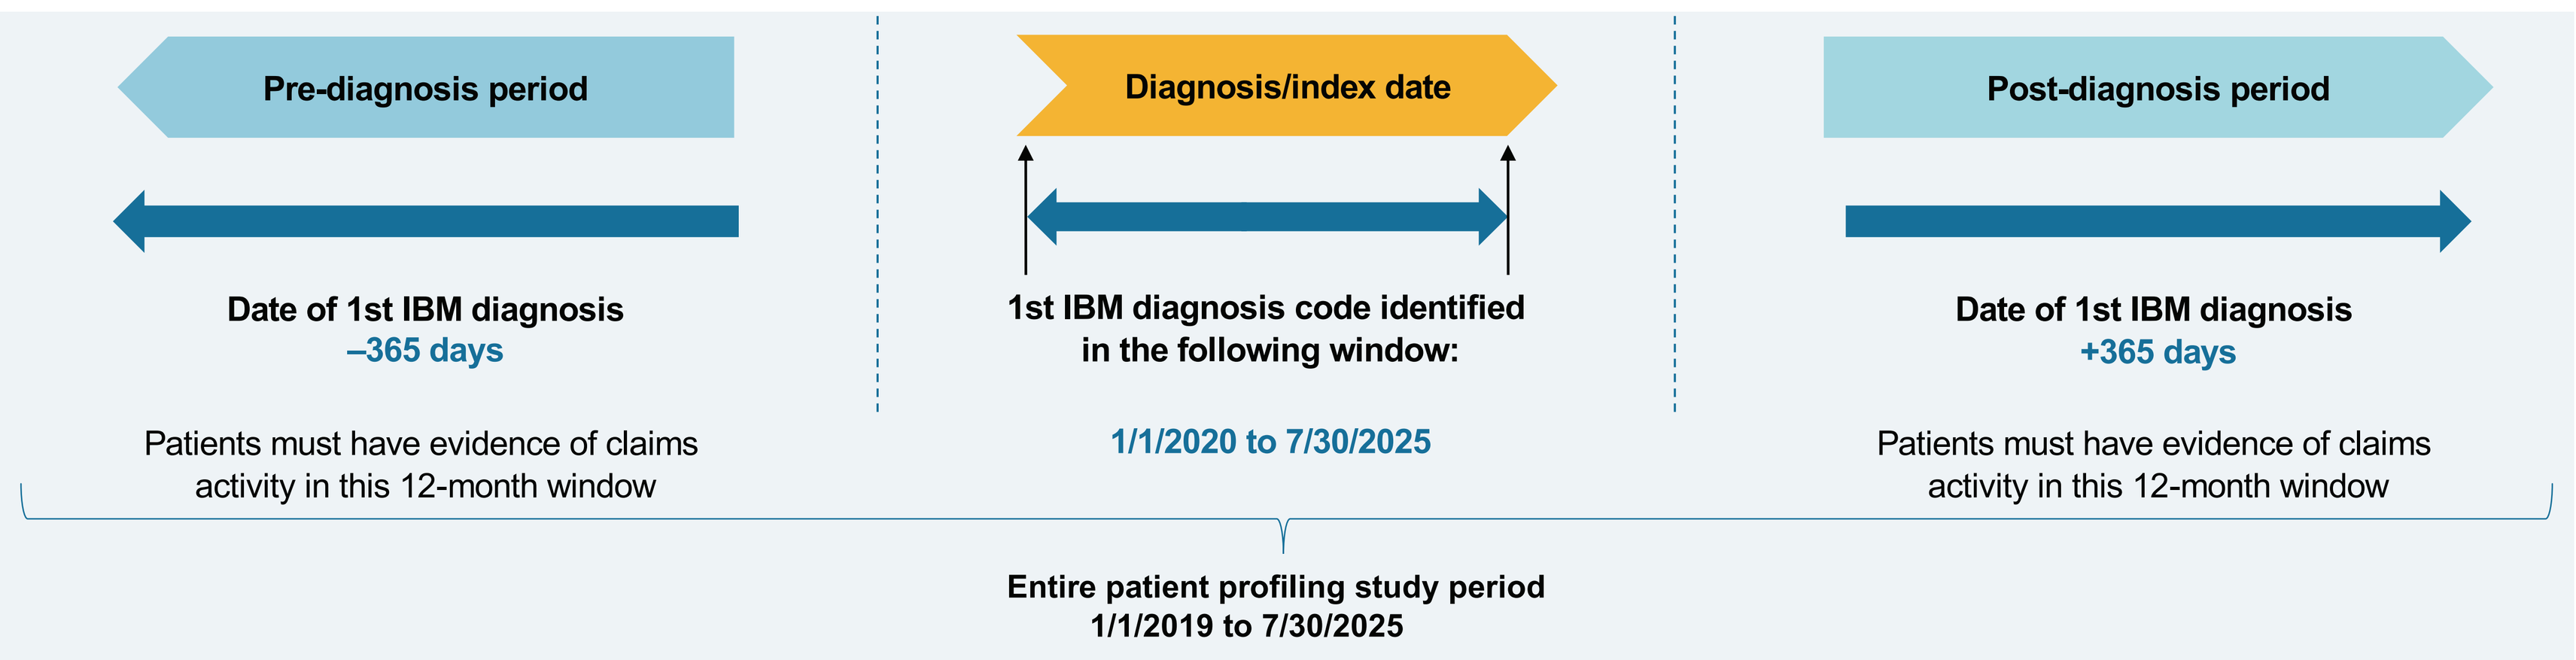
Pre-diagnosis period
Diagnosis/index date
Post-diagnosis period
Date of 1st IBM diagnosis
–365 days
1st IBM diagnosis code identified
in the following window:
Date of 1st IBM diagnosis
+365 days
Patients must have evidence of claims activity in this 12-month window
1/1/2020 to 7/30/2025
Patients must have evidence of claims activity in this 12-month window
Entire patient profiling study period
1/1/2019 to 7/30/2025

IBM, inclusion body myositis.

Machine-learning analyses

- Machine learning was used on an expanded dataset to estimate the number of symptomatic but undiagnosed patients
- Clinical characteristics in the claims data of confirmed diagnosed patients were used to develop a machine learning algorithm to identify an undiagnosed population

Results

Claims analyses

Demographic characteristics

- Data from 3659 patients with IBM and 106,968 matched controls were included in this analysis
- Among patients with IBM, 69% were aged ≥65 years, 60% were male, 67% were white, 41% had commercial insurance coverage, and 40% had Medicare insurance coverage

Diagnosis in patients with IBM

- Median (SD) time from first IBM-related symptom reported to a healthcare practitioner to diagnosis was 2.6 (2.3) years (n=3443 patients)
- Of 41% of patients with IBM who originally received a diagnosis other than IBM, “myositis” was most common (18%), followed by “polymyositis” (8%; **Figure 2**)

Figure 2. Differential diagnosis patterns in patients with IBM

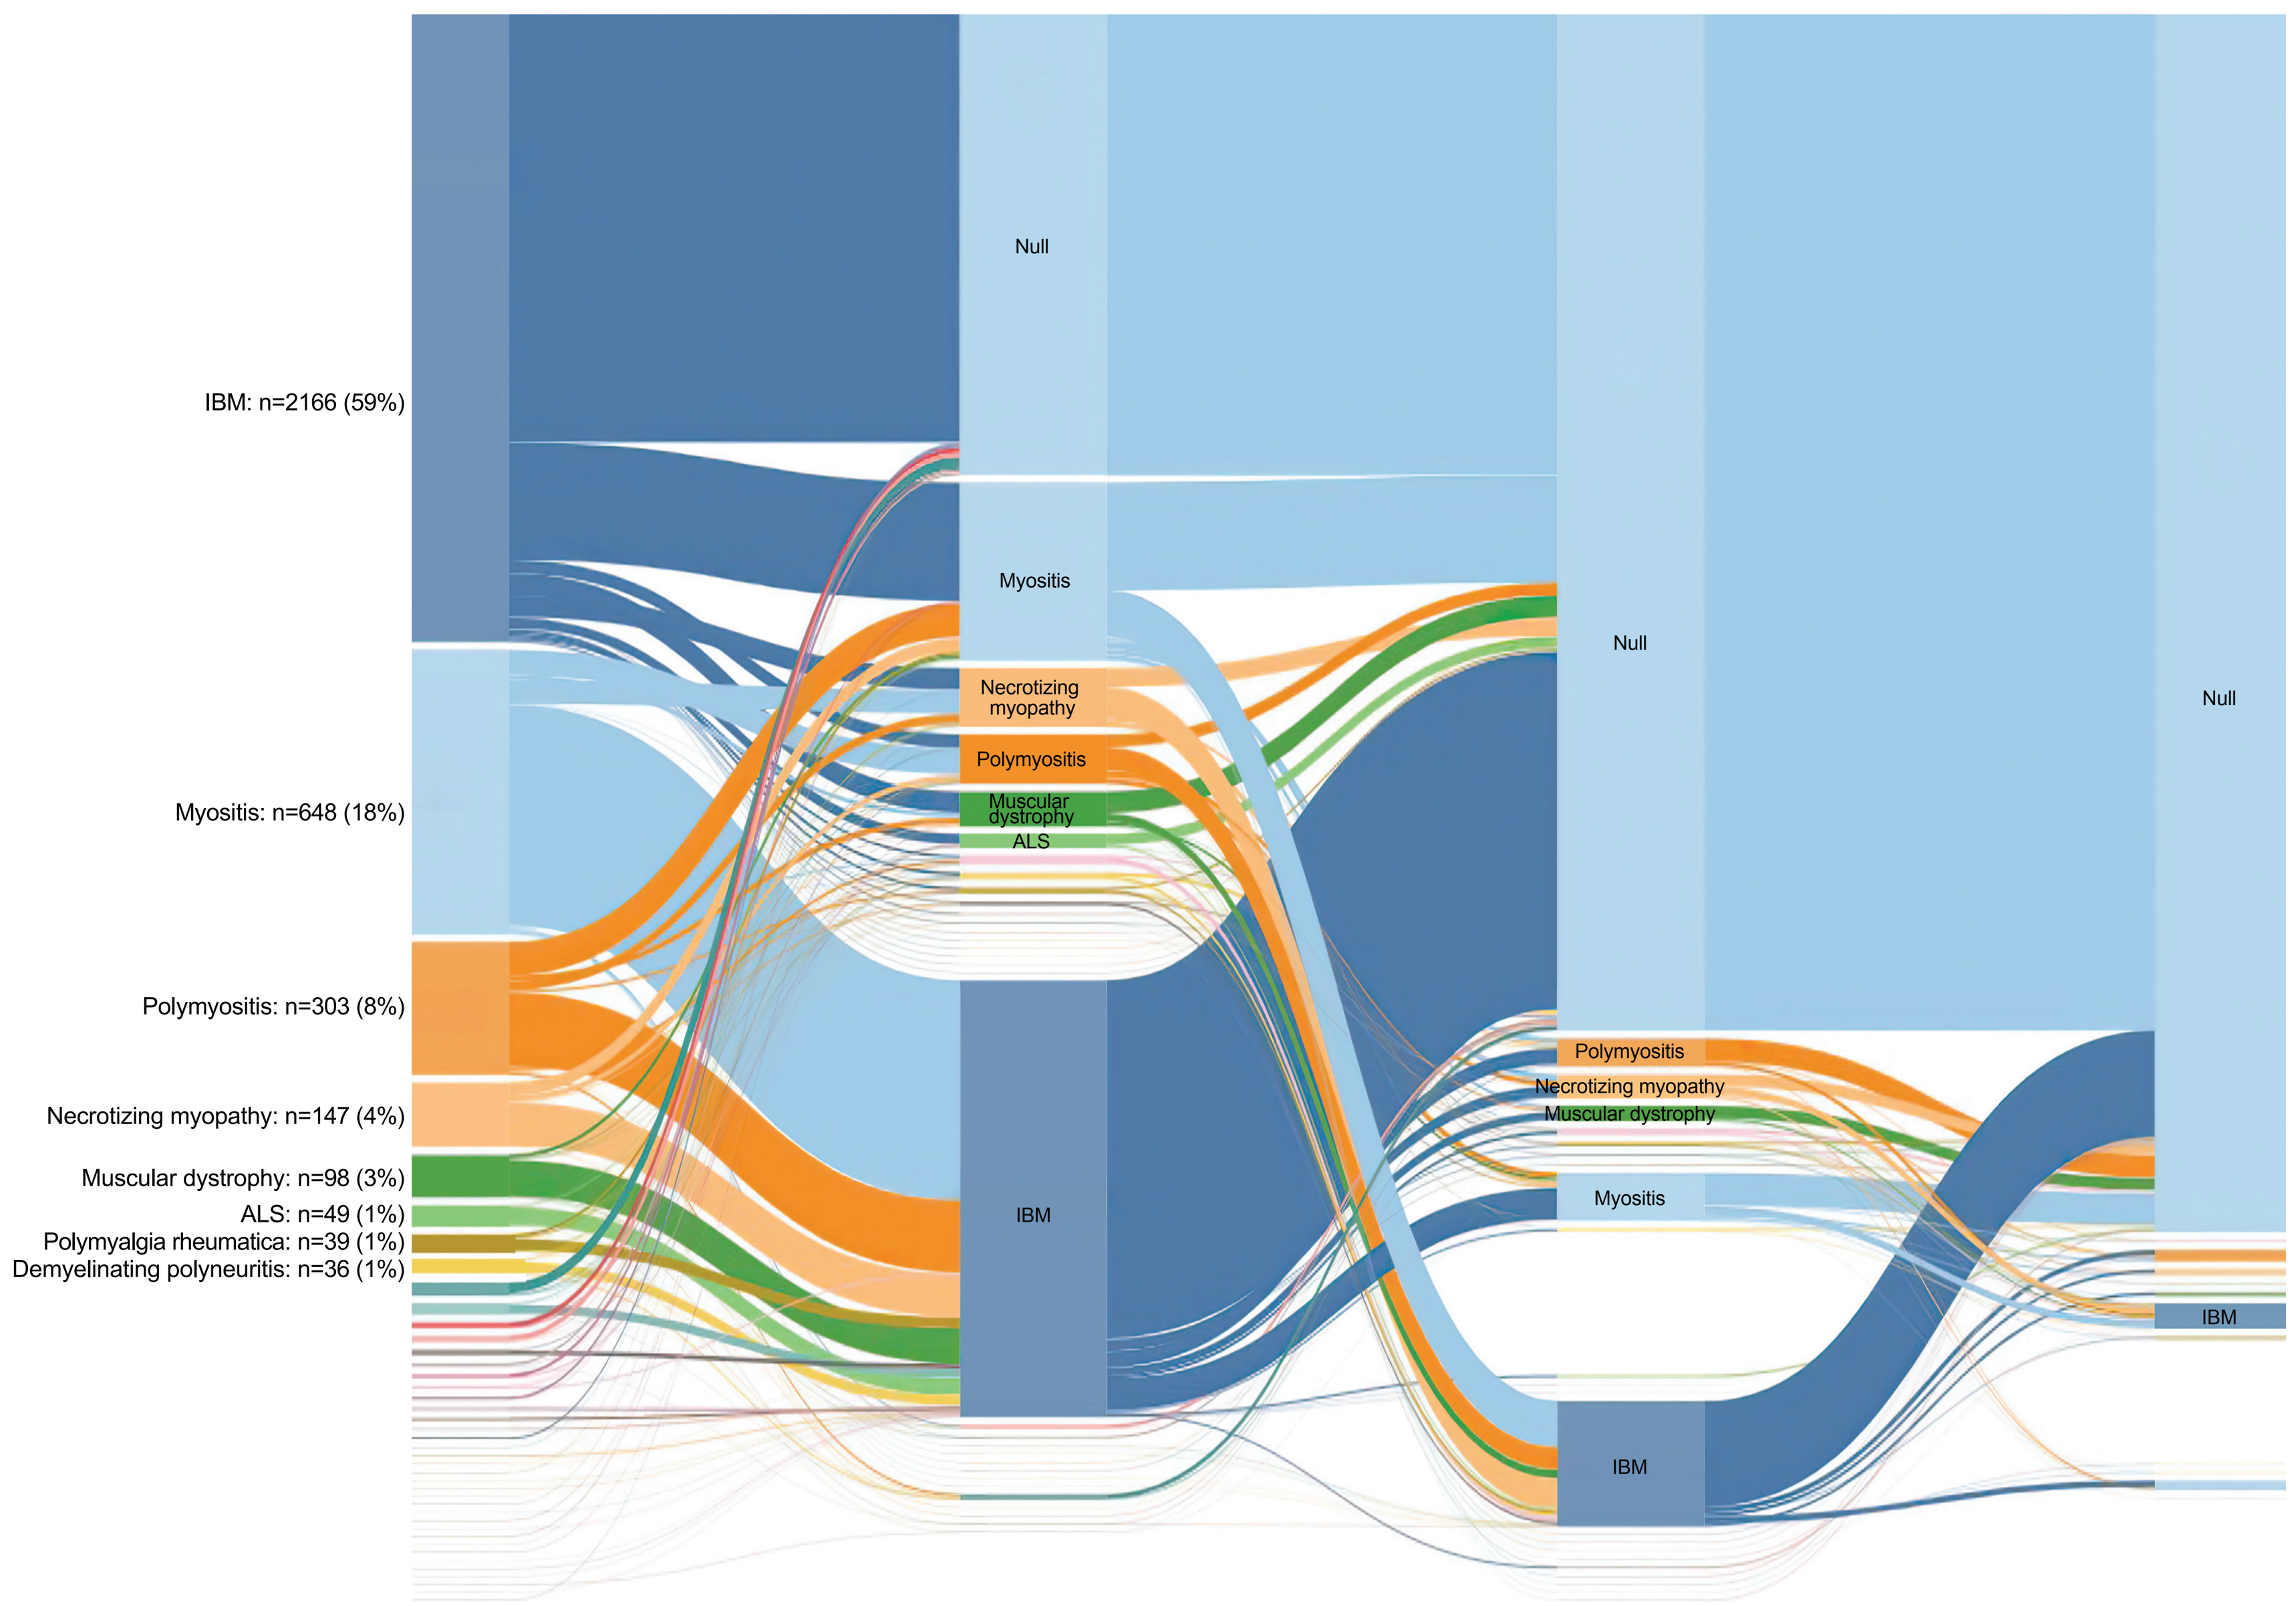
IBM: n=2166 (59%)
Myositis: n=648 (18%)
Polymyositis: n=303 (8%)
Necrotizing myopathy: n=147 (4%)
Muscular dystrophy: n=98 (3%)
ALS: n=49 (1%)
Polymyalgia rheumatica: n=39 (1%)
Demyelinating polyneuritis: n=36 (1%)
Null
Myositis
Necrotizing myopathy
Polymyositis
Muscular dystrophy
ALS
IBM
Null
Polymyositis
Necrotizing myopathy
Muscular dystrophy
Myositis
IBM
Null
IBM

ALS, amyotrophic lateral sclerosis; IBM, inclusion body myositis.

Comorbidities after diagnosis in patients with IBM

- Cardiac conditions, musculoskeletal pain, impaired mobility, and general pain were the most common comorbidities in the IBM cohort postdiagnosis and were all more common in the IBM cohort than in controls (**Figure 3A**)
 - The most common cardiac comorbidities were captured as heart disease (14% in IBM vs 10% in controls), atherosclerosis (12% vs 8%), and atrial fibrillation (9% vs 6%); there was minimal difference in reported rates of cardiomyopathy versus controls (**Figure 3B**)
- Immune-mediated disorders were more common in patients with IBM than in controls postdiagnosis, including Sjögren syndrome (16.1 times [x] more common), ankylosing spondylitis (7.3x), lupus (7.2x), polyneuropathy (3.9x), rheumatoid arthritis (2.6x), multiple sclerosis (2.2x), and psoriatic arthritis (2.1x)
- Several hematologic malignancies were also more common in patients with IBM than in controls postdiagnosis, including “other lymphoid leukemia” (52.0x), multiple myeloma (2.4x), and non-Hodgkin lymphoma (2.1x)
- Incidences of solid tumors and neurodegenerative diseases were similar between groups

Figure 3. (A) Prevalence of the most common comorbidities and (B) specific cardiac comorbidities in patients with IBM versus matched controls postdiagnosis

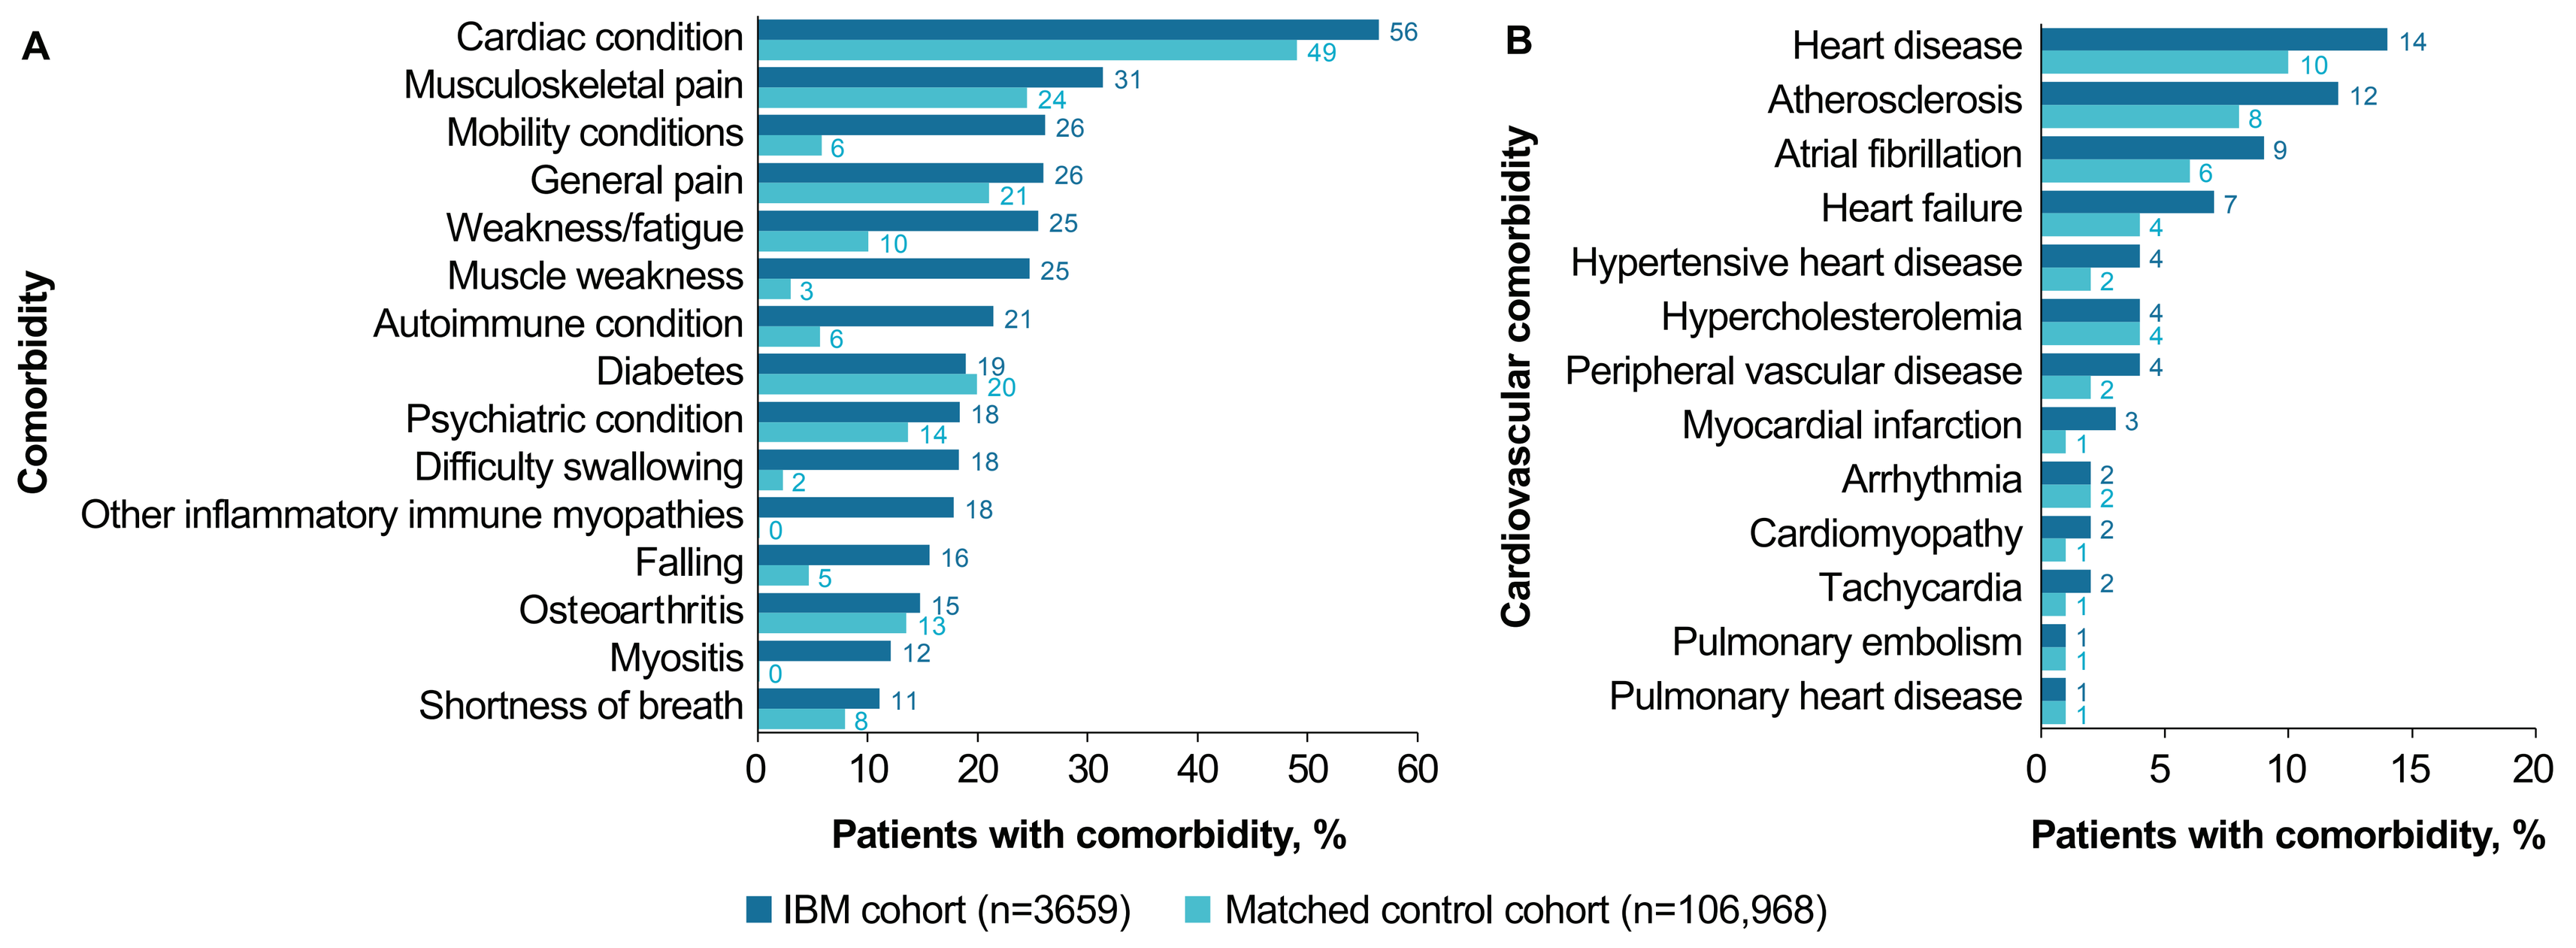
A
Comorbidity
Cardiac condition 56 49
Musculoskeletal pain 31 24
Mobility conditions 26 6
General pain 26 21
Weakness/fatigue 25 10
Muscle weakness 25 3
Autoimmune condition 21 6
Diabetes 19 20
Psychiatric condition 18 14
Difficulty swallowing 18 2
Other inflammatory immune myopathies 18 0
Falling 16 5
Osteoarthritis 15 13
Myositis 12 0
Shortness of breath 11 8
0 10 20 30 40 50 60
Patients with comorbidity, %
B
Cardiovascular comorbidity
Heart disease 14 10
Atherosclerosis 12 8
Atrial fibrillation 9 6
Heart failure 7 4
Hypertensive heart disease 4 2
Hypercholesterolemia 4 4
Peripheral vascular disease 4 2
Myocardial infarction 3 1
Arrhythmia 2 2
Cardiomyopathy 2 1
Tachycardia 2 1
Pulmonary embolism 1 1
Pulmonary heart disease 1 1
0 5 10 15 20
Patients with comorbidity, %
IBM cohort (n=3659)
Matched control cohort (n=106,968)

IBM, inclusion body myositis.

HCRU in patients with IBM after diagnosis

- Patients with IBM had more outpatient office and hospital visits, emergency room visits, inpatient hospital visits, and use of skilled nursing services and hospice care compared with controls in the 12 months after diagnosis (**Figure 4**)

Figure 4. Healthcare settings visited during the first 12 months after diagnosis in patients with IBM versus matched controls

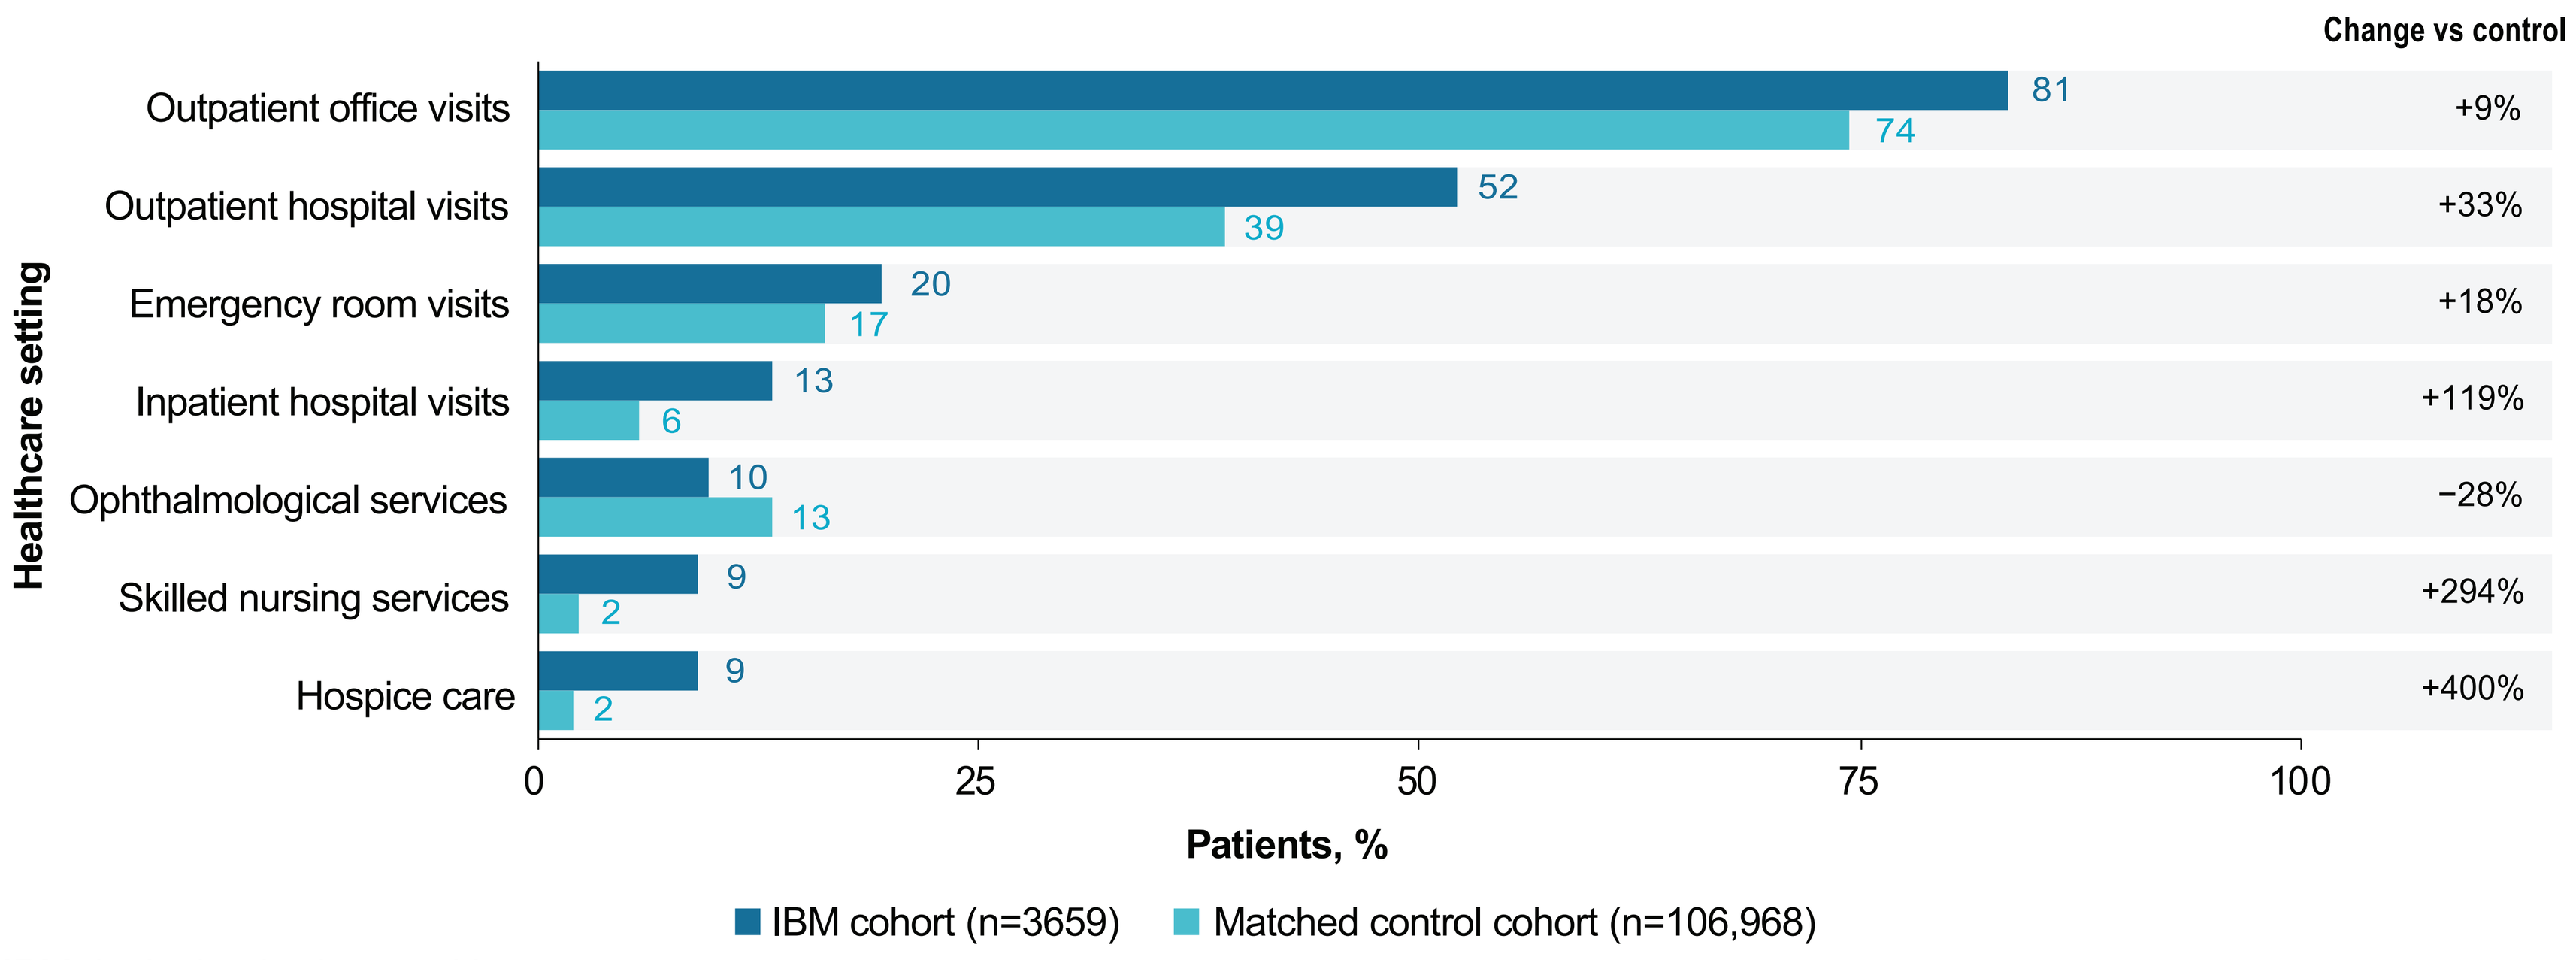
Change vs control
Healthcare setting
Outpatient office visits 81 74 +9%
Outpatient hospital visits 52 39 +33%
Emergency room visits 20 17 +18%
Inpatient hospital visits 13 6 +119%
Ophthalmological services 10 13 −28%
Skilled nursing services 9 2 +294%
Hospice care 9 2 +400%
0 25 50 75 100
Patients, %
IBM cohort (n=3659)
Matched control cohort (n=106,968)

IBM, inclusion body myositis.

Mortality rates in patients with IBM

- All-cause mortality rates in 2020–2025 were significantly increased in patients with IBM compared with controls for several age groups (**Figure 5**); SMRs in the age ranges of 38−47, 48−57, 58−67, and 68+ years ranged from 1.76−8.55 (all *P*<0.05)
 - A subgroup analysis of mortality by age groups, sex, and race showed similar patterns of increased mortality with IBM compared with controls

Figure 5. All-cause mortality rates in 2020−2025 by age in the IBM cohort versus matched controls

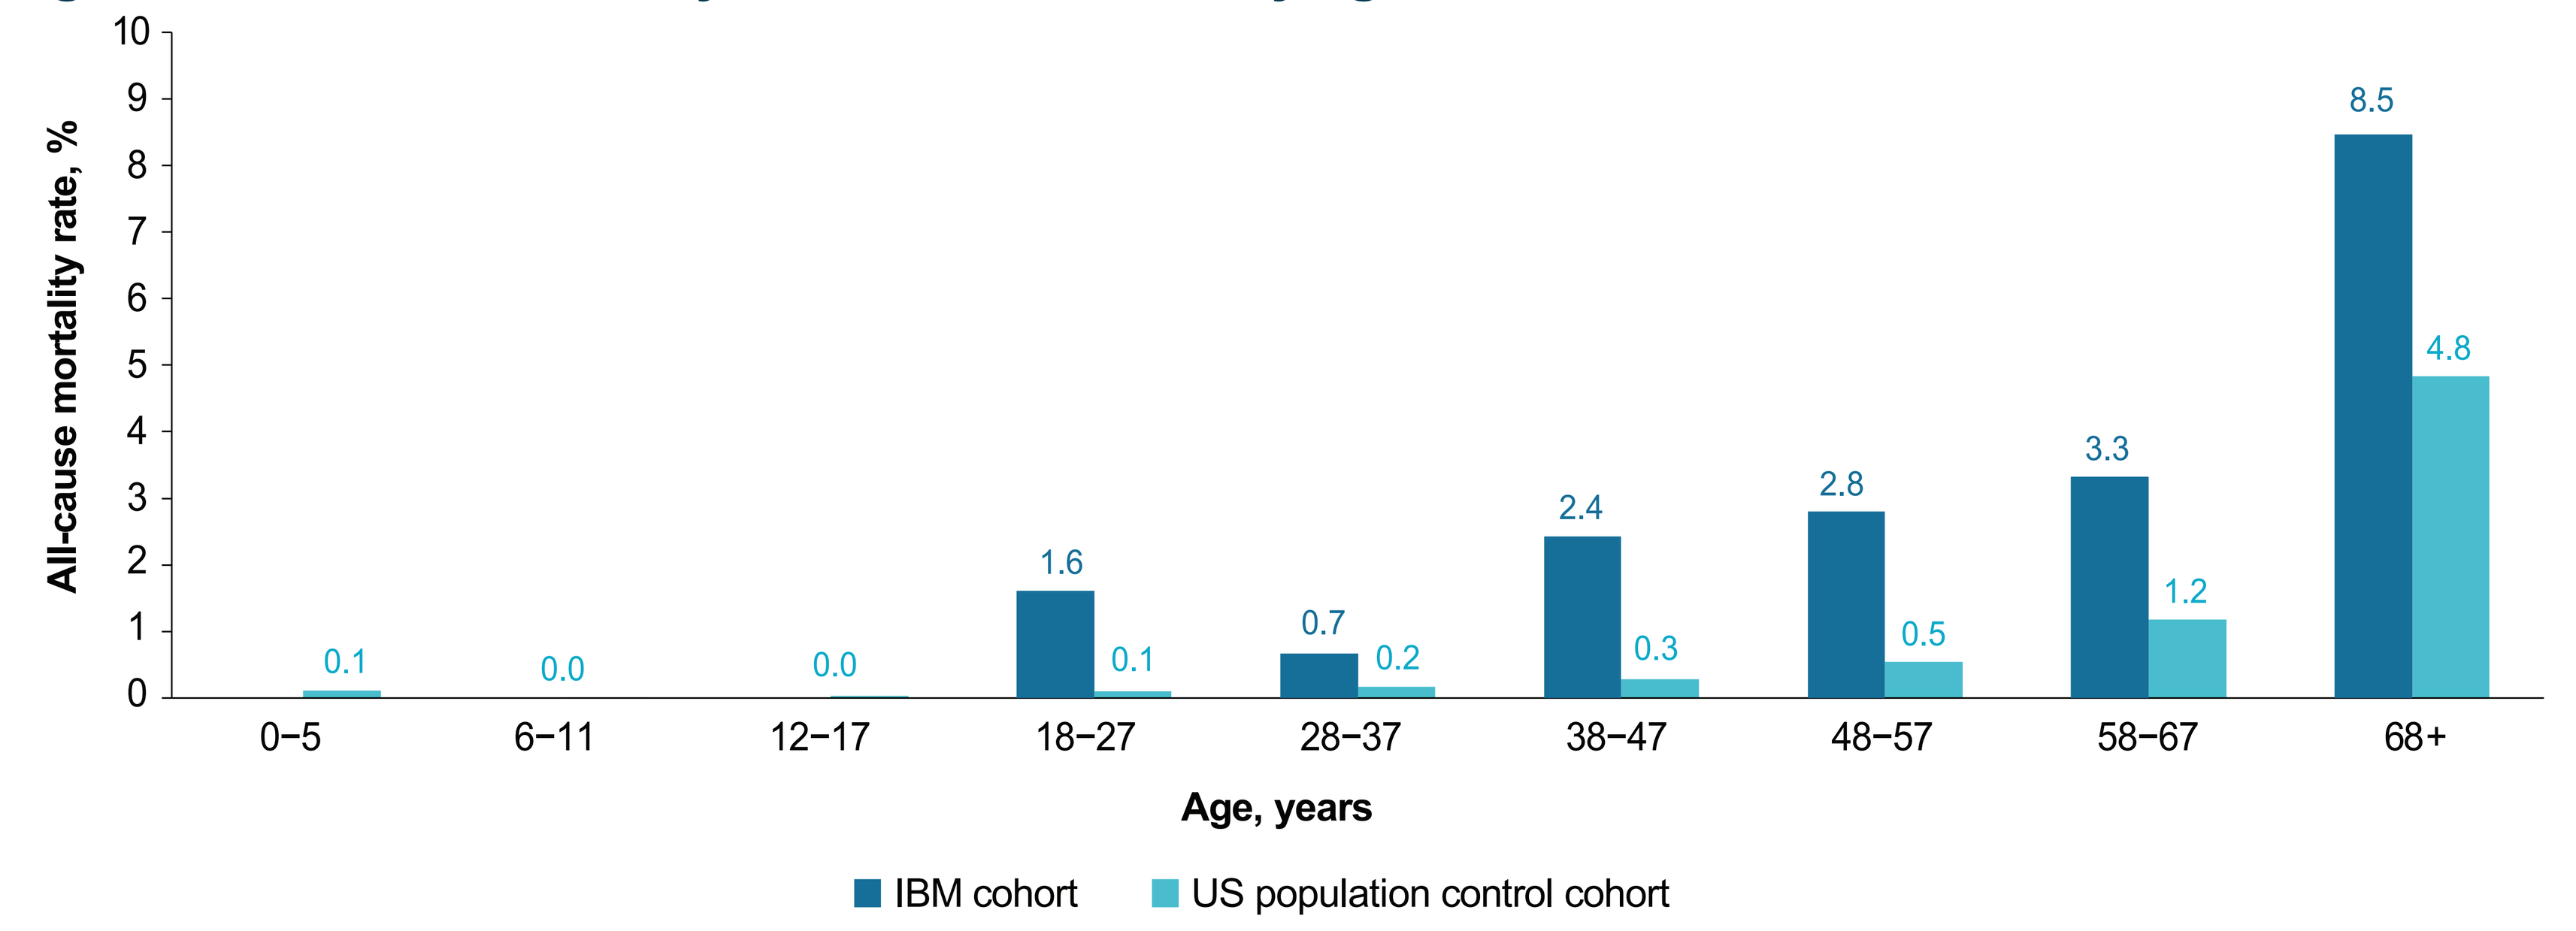
All-cause mortality rate, %
0 1 2 3 4 5 6 7 8 9 10
0−5 0.1
6−11 0.0
12−17 0.0
18−27 1.6 0.1
28−37 0.7 0.2
38−47 2.4 0.3
48−57 2.8 0.5
58−67 3.3 1.2
68+ 8.5 4.8
Age, years
IBM cohort
US population control cohort

Patients with IBM all-cause mortality rate (open claims data) = yearly deceased patients with IBM / yearly living patients with IBM; US population all-cause mortality rate (CDC data) = yearly deceased in US / yearly US population. CDC, US Centers for Disease Control and Prevention; IBM, inclusion body myositis.

Machine-learning analyses

- Depending on the specificity used in the machine-learning model, for every patient diagnosed with IBM, there are 1.2–1.9 patients who have symptoms of IBM but lack a formal diagnosis

Conclusions

- A claims analysis showed that patients with IBM experience increased HCRU and earlier mortality compared with a matched control cohort
- A machine-learning analysis suggested that for every patient diagnosed with IBM in the US, there is at least 1 patient who is symptomatic and undiagnosed
- Earlier recognition of IBM is needed to provide timely and appropriate care

Presented at the 6th Global Conference on Myositis (GCOM)
March 23–26, 2026 | Lisbon, Portugal

Disclosures

REM: Consultant to Abcuro, Inc.; **KT:** Employee of Abcuro, Inc.; **JW** and **CK:** Employees of Real Chemistry; **EN:** Received funding for meeting attendance/panelist, or advisory board participation from Medscape, Johnson & Johnson, and Expert Connect; received clinical trial support from Cabaletta, Abcuro, and Fulcrum; served as a site PI for ABC008.

Acknowledgments

This study was sponsored by Abcuro, Inc. Medical writing and editorial assistance were provided by Peloton Advantage, LLC, an OPEN Health company, and were funded by Abcuro, Inc.

References

1. Greenberg SA. *Nat Rev Rheumatol*. 2019;15:257-72.
2. Greenberg SA, et al. *Brain*. 2019;142:2590-604.
3. Lindgren U, et al. *Ann Neurol*. 2022;92:201-12.
4. Paltiel AD, et al. *Muscle Nerve*. 2015;52:527-33.
5. Capkun G, et al. *Muscle Nerve*. 2017;56:861-7.
6. Shelly S, et al. *Neurology*. 2021;96:e2653-e61.
7. Machado P, et al. *Curr Opin Rheumatol*. 2013;25:763-71.
8. Naddaf E, et al. *Rheumatology (Oxford)*. 2022;61:2016-24.